In vitro Functional Interrogation of Viable Circulating Tumor Associated Cells (C-TACs) for Evaluating Platin Resistance.

Stefan Schuster[1], Sanket Patil[2], Darshana Patil[2], Vineet Datta[2], Ajay Srinivasan[2], Rajan Datar[2], Dadasaheb Akolkar[2].

[1]Datar Cancer Genetics Europe GmbH, Germany
[2]Datar Cancer Genetics Limited, India;

Conflict of Interest:
Datar Cancer Genetics Limited and Datar Cancer Genetics Europe GmbH offer commercial services in the domain of oncology.

BACKGROUND

- Innate and acquired platin resistance lead to failure of first as well as subsequent lines of treatment,
- Non-invasive real-time monitoring for platin resistance can guide precision therapy management,
- Longitudinal chemoresistance profiling (CRP) of tumor derived cells (TDCs) is unsuitable since it necessitates repeated invasive biopsies,
- Conventional methods for harvesting Circulating Tumor Cells (CTCs) such as EpCAM-capture or microfluidic devices have low yields, due to which CRP of CTCs is not feasible.

RATIONALE

- Circulating Tumor Associated Cells (C-TACs) are EpCAM+, CK+, CD45± cells of tumorigenic origin, in peripheral blood,
- Non-tumorigenic cells in peripheral blood have functional apoptotic mechanism, but C-TACs are resistant to apoptosis,
- An epigenetically acting stabilizing process can eliminate normal cells and confer survival privilege on apoptosis-resistant C-TACs,
- Sufficient C-TACs can be enriched by such negative selection and harvested for CRP.

APPROACH

- 15 mL peripheral blood was obtained from 708 patients with breast, colorectal, lung, ovarian and pancreatic cancers,
- Viable C-TACs enriched and harvested from PBMC fraction,
- C-TACs treated with various platinum agents and apoptotic fraction estimated to determine resistance or sensitivity,
- CRP to determine platin-resistance concordance between C-TACs and TDCs (Arm 1), acquired platin-resistance (Arm 2) and innate platin resistance (Arm 3).

STUDY DESIGN

Figure 1. The study was based on a cohort of 709 patients

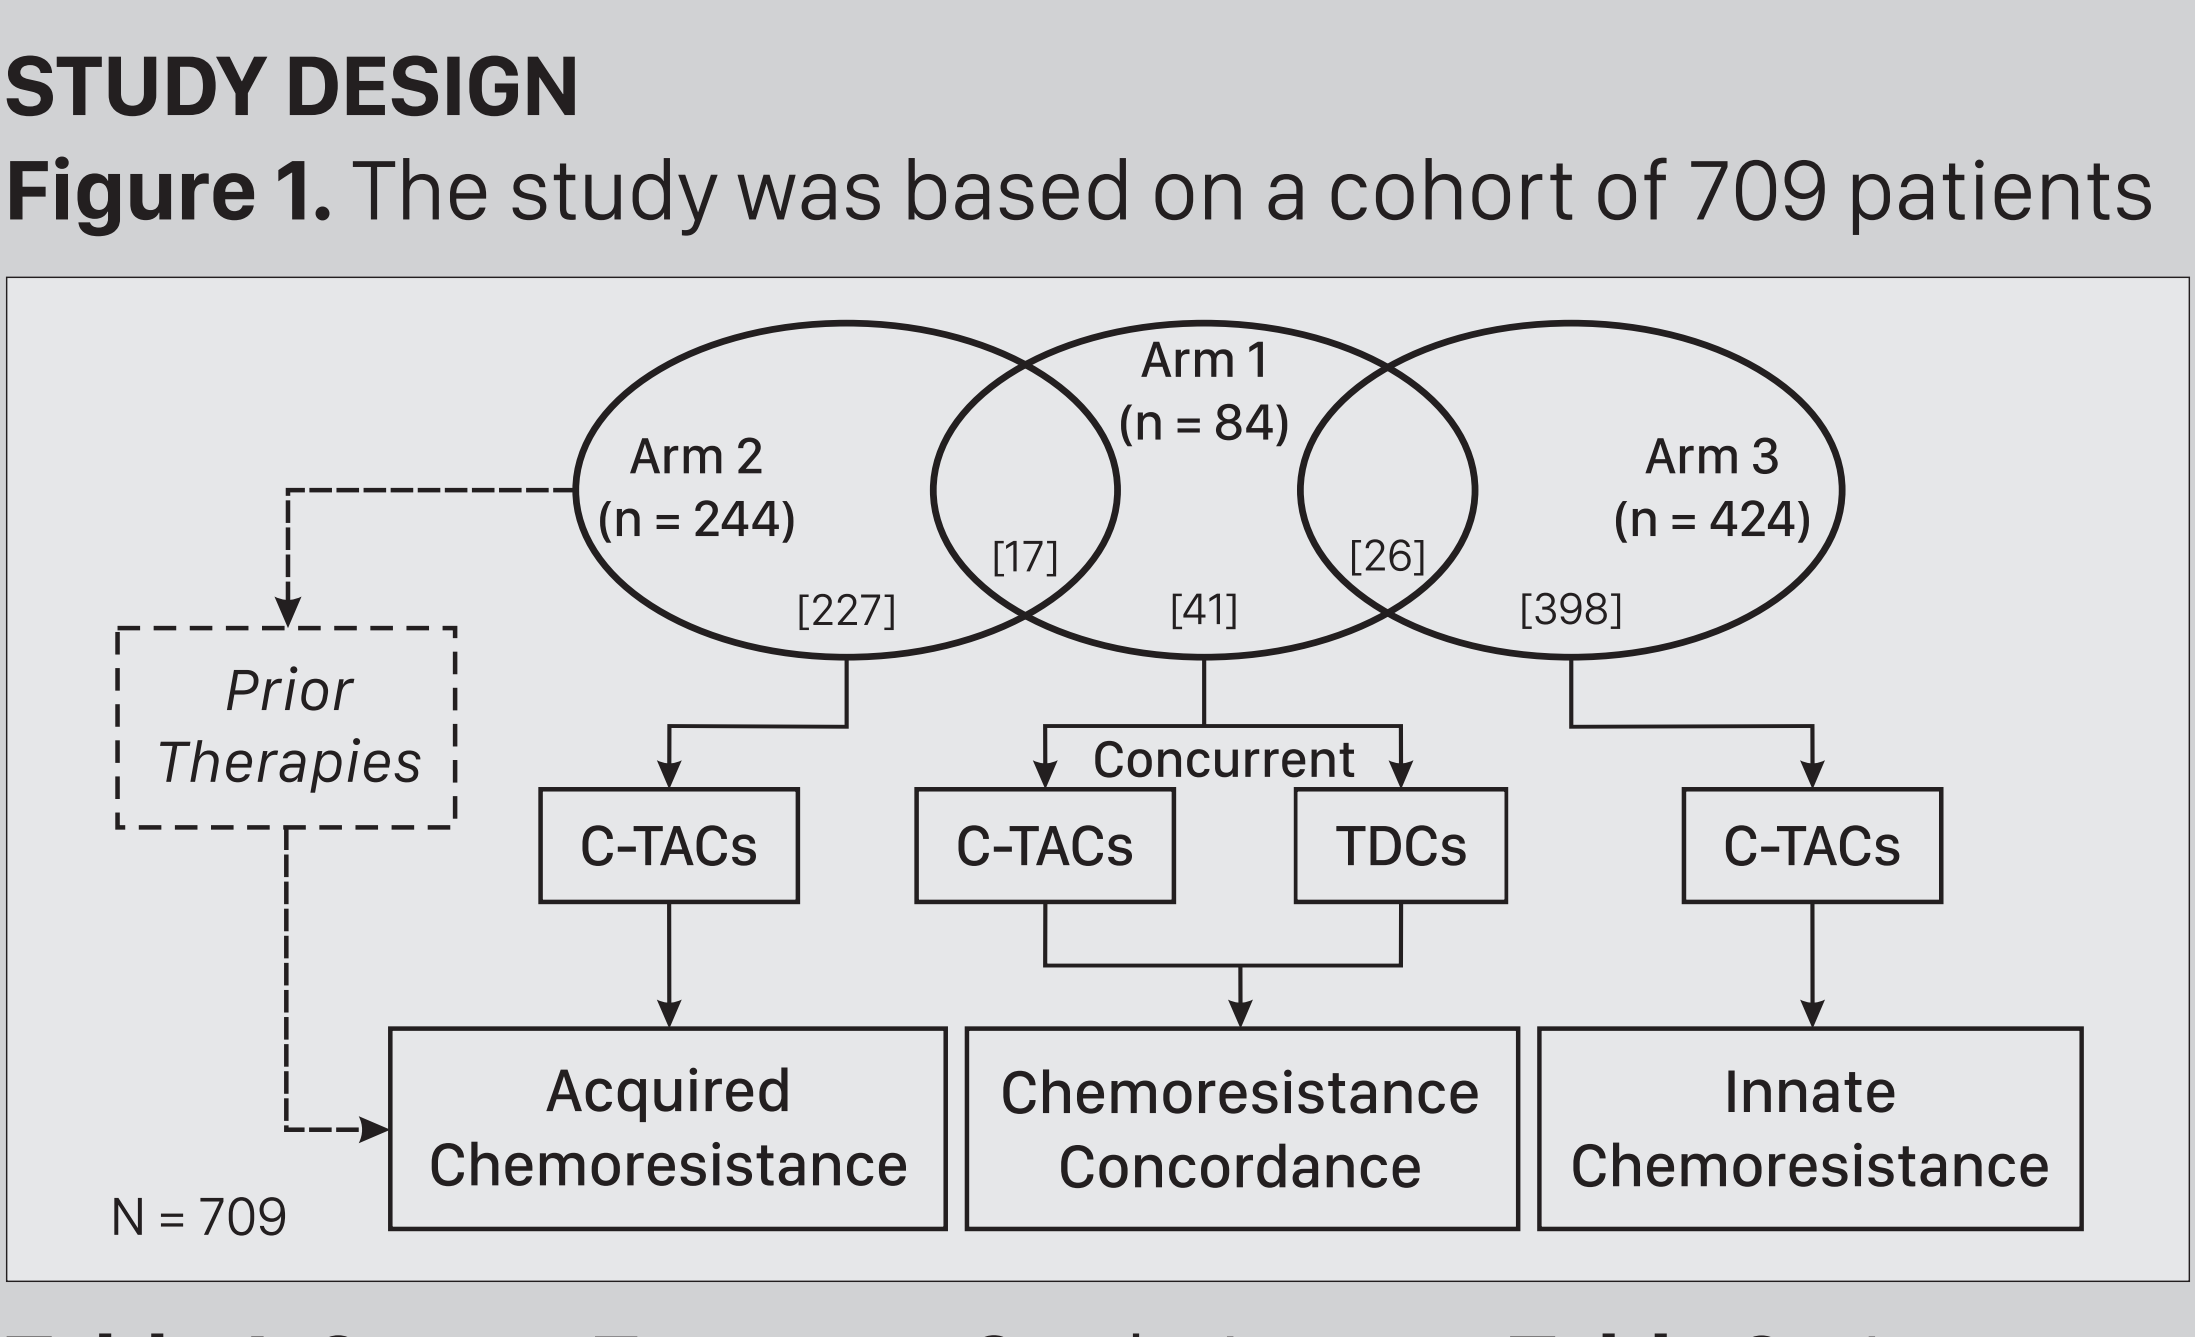
Arm 1
(n = 84)
Arm 2
(n = 244)
Arm 3
(n = 424)
[227]
[17]
[41]
[26]
[398]
Prior Therapies
Concurrent
C-TACs
C-TACs
TDCs
C-TACs
Acquired Chemoresistance
Chemoresistance Concordance
Innate Chemoresistance
N = 709

Table 1. Cancer Types per Study Arm.

Cancer Type	Arm 1	Arm 2	Arm 3
Breast	46	127	218
Colorectal	13	43	55
Lung	9	25	102
Ovarian	14	37	20
Pancreatic	2	12	29
Sub-Total	84	244	424

Table 2. Age.

Age	Years
Minimum	14
Maximum	95
Median	55

Table 3. Gender.

Gender	# (%)
Male	192 (27%)
Female	517 (73%)

C-TACS YIELD AND CHARACTERIZATION

Figure 2. Sufficient C-TACs were enriched and harvested across all cancer types and irrespective of treatment status (N: Therapy Naïve, T: Pre-treated). C-TACs yield appeared to be higher in therapy naïve patients.

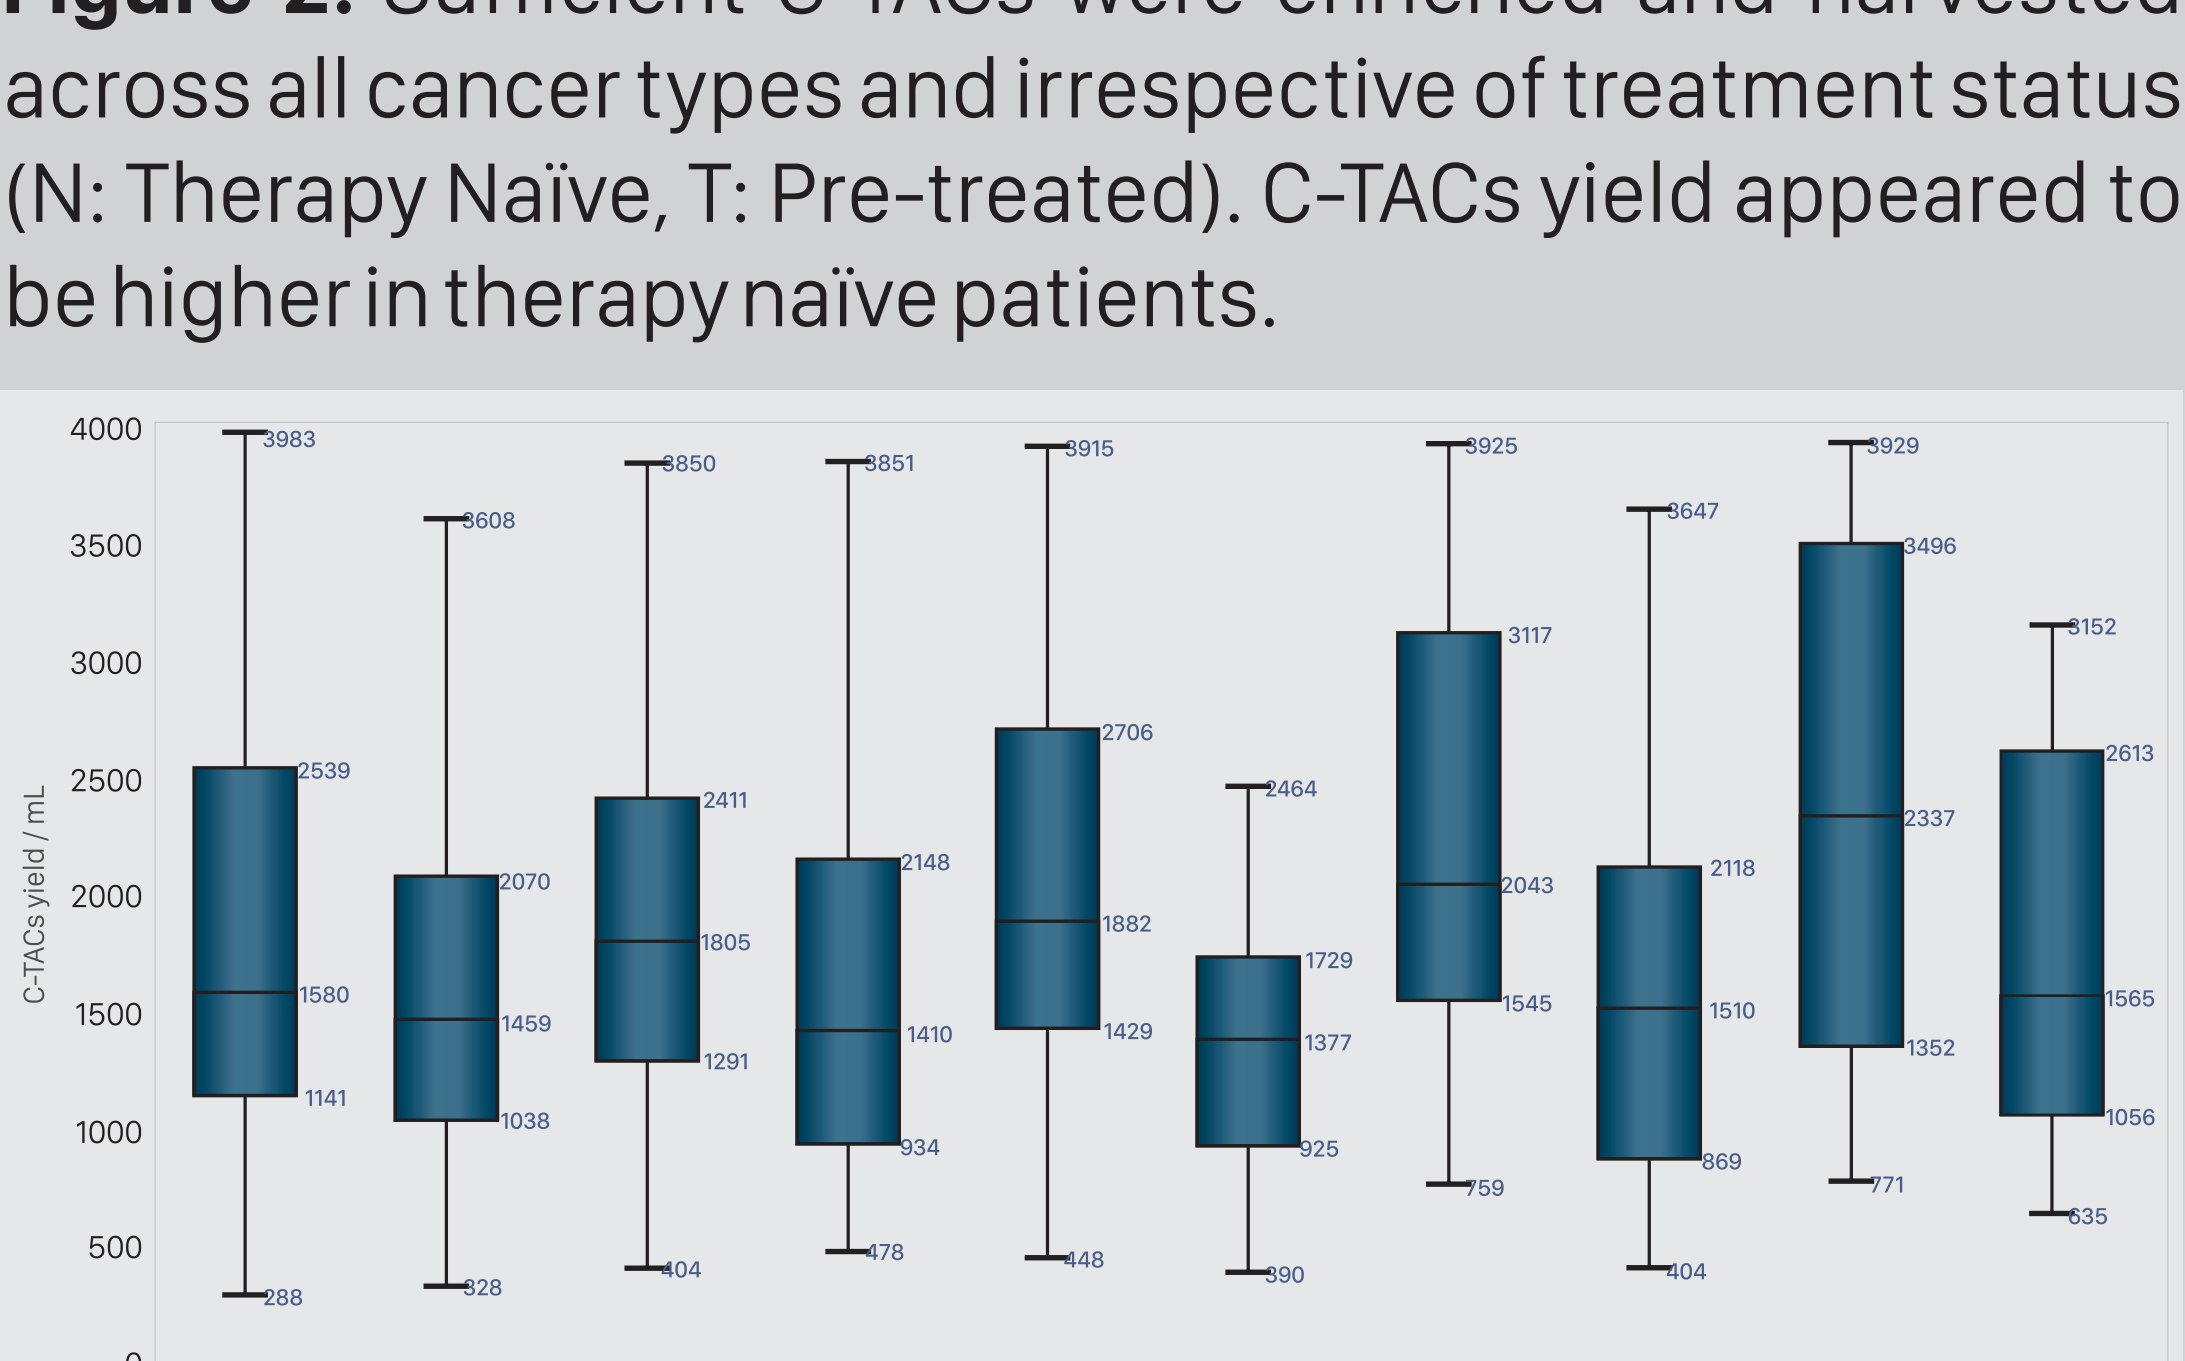
C-TACs yield / mL
Breast (N)
Breast (T)
Colorectal (N)
Colorectal (T)
Lung (N)
Lung (T)
Ovarian (N)
Ovarian (T)
Pancreatic (N)
Pancreatic (T)

Figure 3. Representative images of Immunocytochemistry profiles of C-TACs from Ca Breast (A-E) and Ca Colon (F-J). A, F: DAPI nucleic acid stain; B, G: anti-pan-CK-FITC; C, H: anti-EpCAM-PE; D, I: anti-CD45-Cy5; E, J: overlaid images. C-TACs are EpCAM+, pan-CK+ and CD45± cells. White bar = 50 µm.

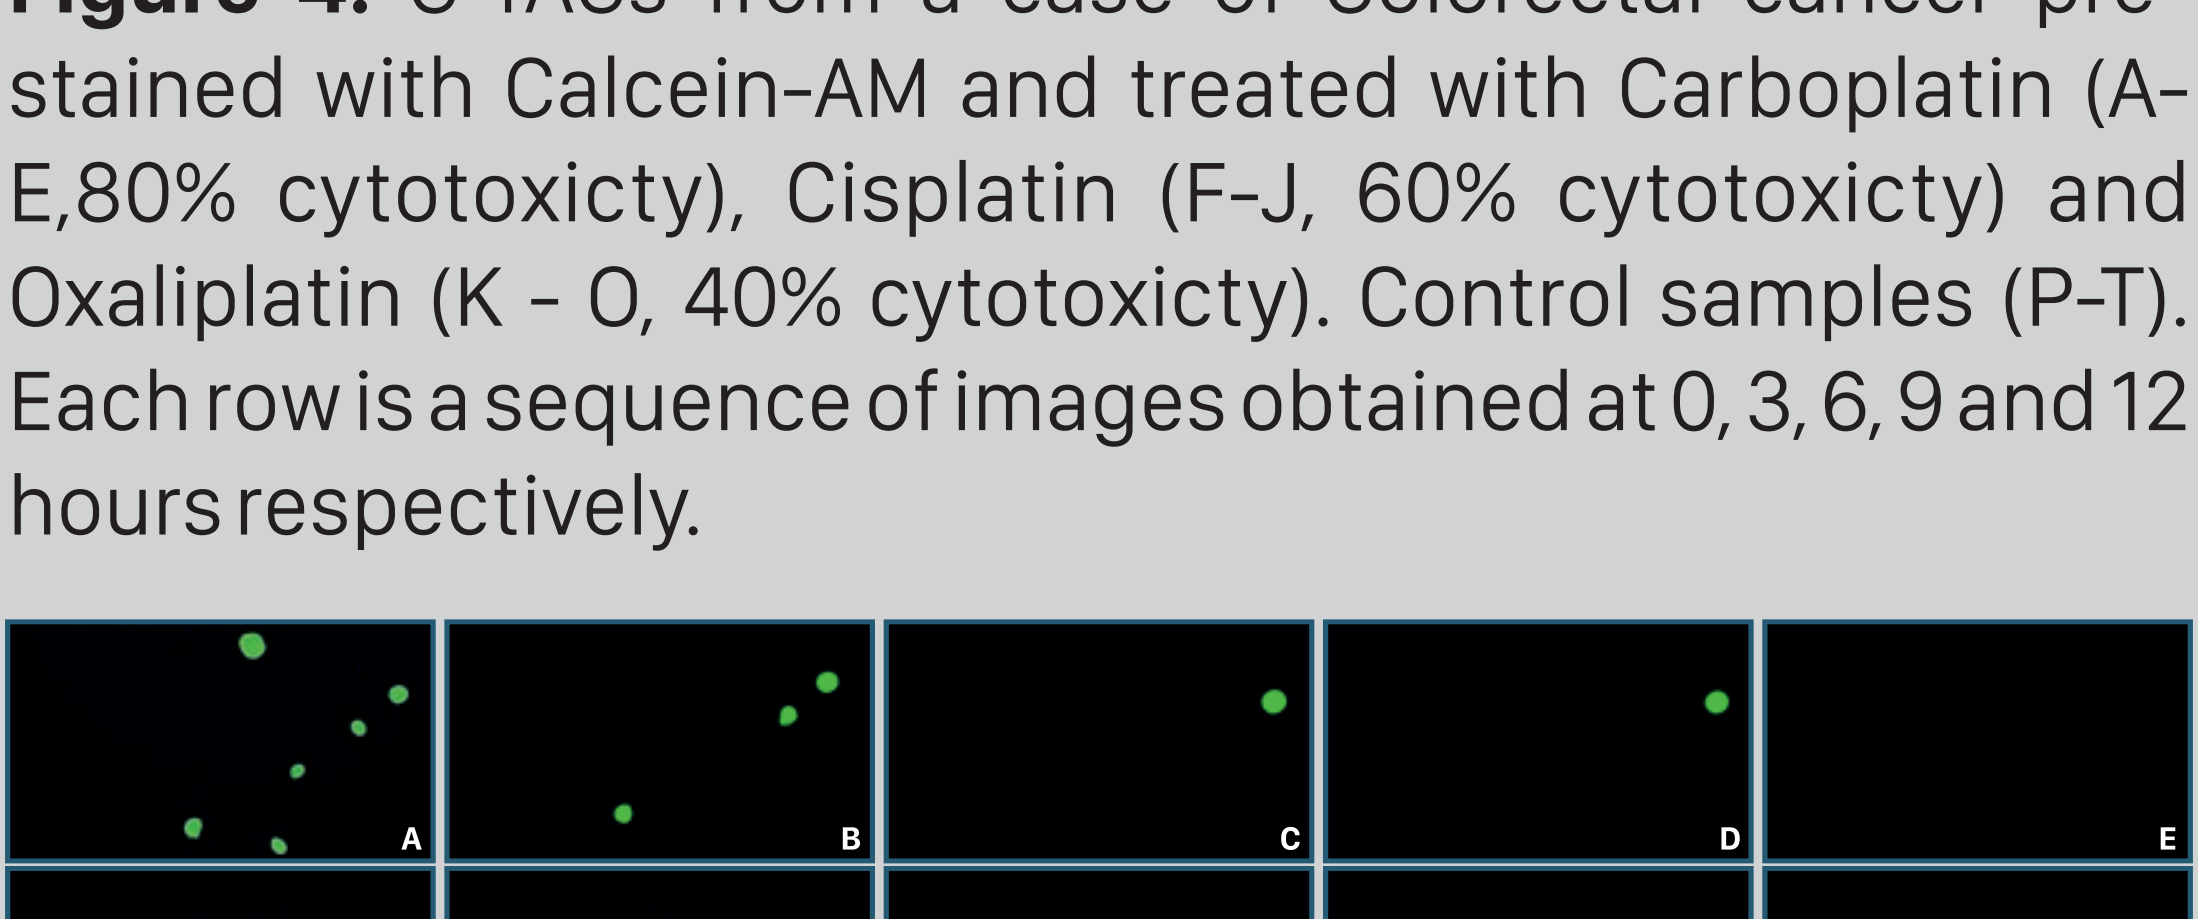

IN VITRO CHEMORESISTANCE PROFILING OF C-TACs

Figure 4. C-TACs from a case of Colorectal cancer pre-stained with Calcein-AM and treated with Carboplatin (A-E, 80% cytotoxicty), Cisplatin (F-J, 60% cytotoxicty) and Oxaliplatin (K - O, 40% cytotoxicty). Control samples (P-T). Each row is a sequence of images obtained at 0, 3, 6, 9 and 12 hours respectively.

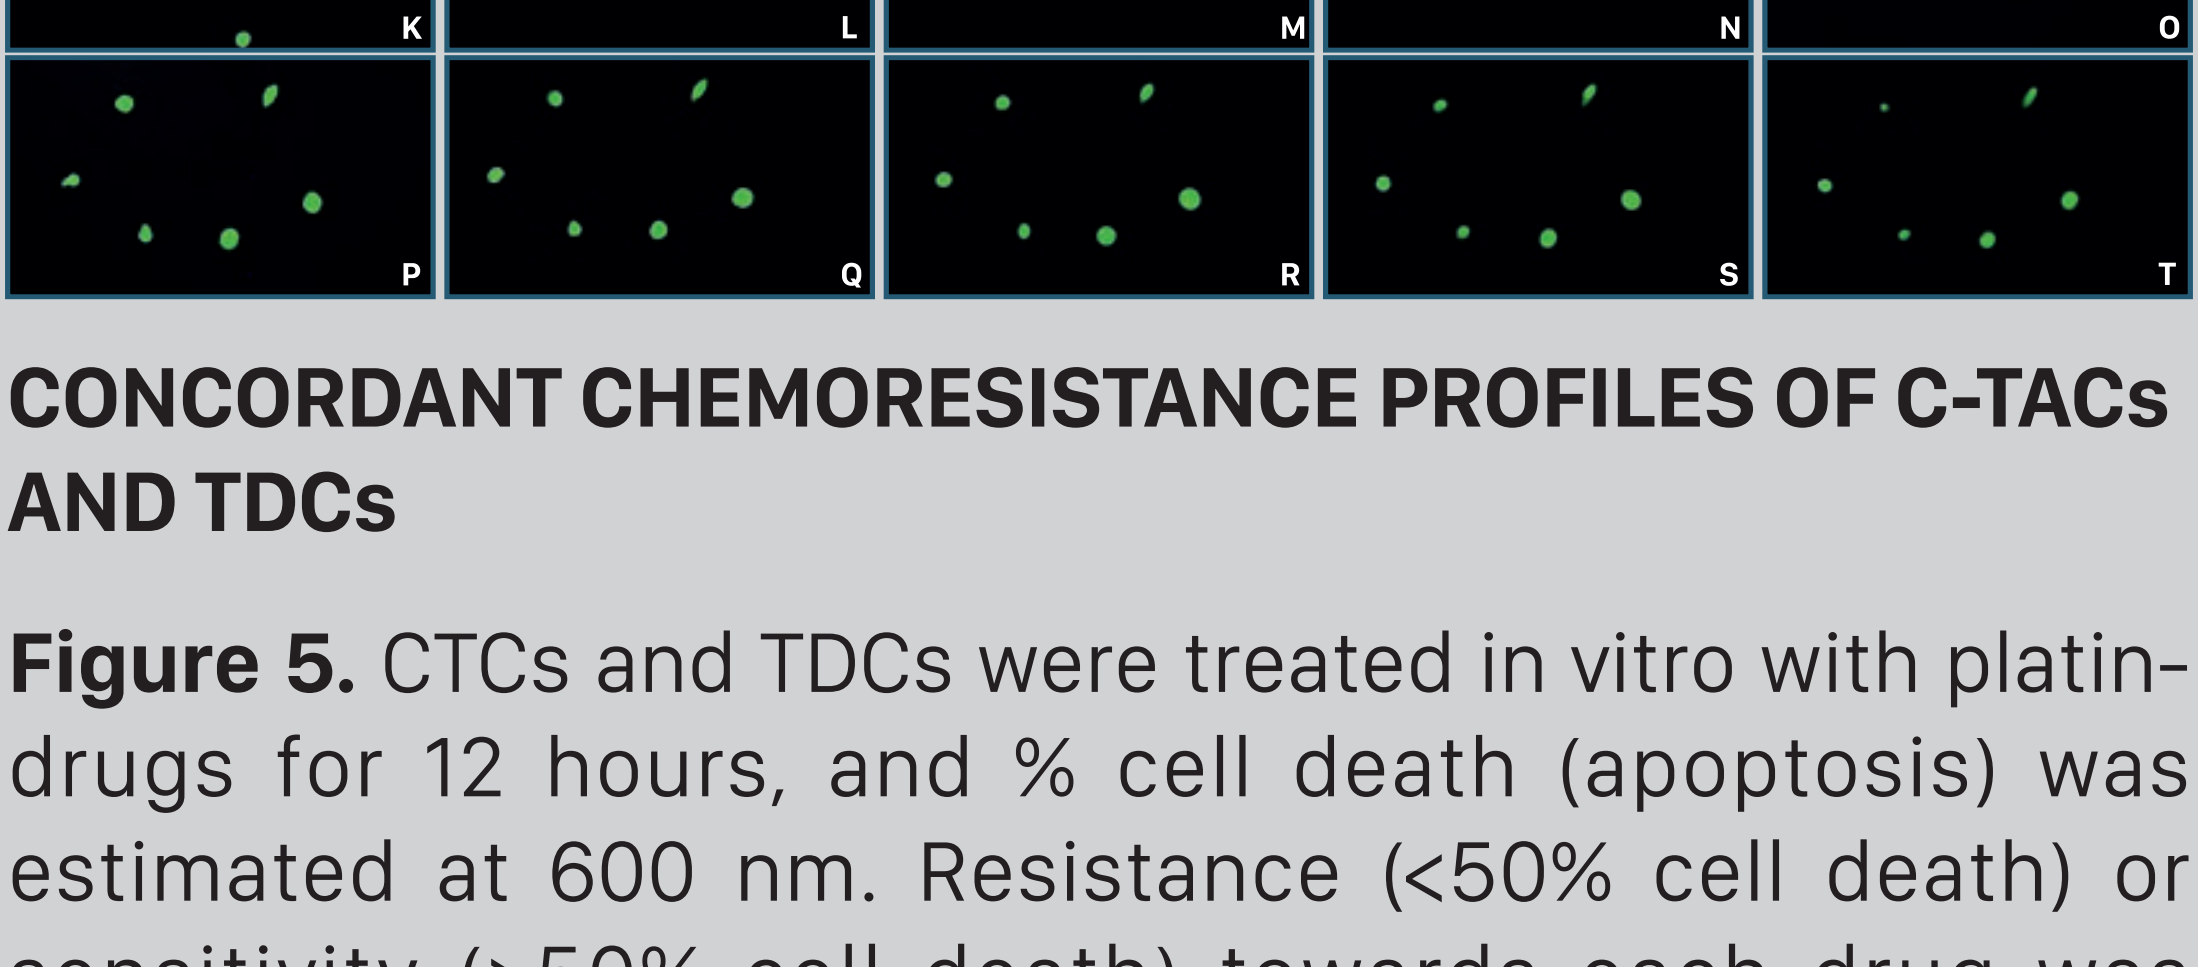

CONCORDANT CHEMORESISTANCE PROFILES OF C-TACs AND TDCs

Figure 5. CTCs and TDCs were treated in vitro with platin-drugs for 12 hours, and % cell death (apoptosis) was estimated at 600 nm. Resistance (<50% cell death) or sensitivity (>50% cell death) towards each drug was determined. Significant concordance was observed between paired CTCs and TDCs for resistance or sensitivity towards platin drugs.

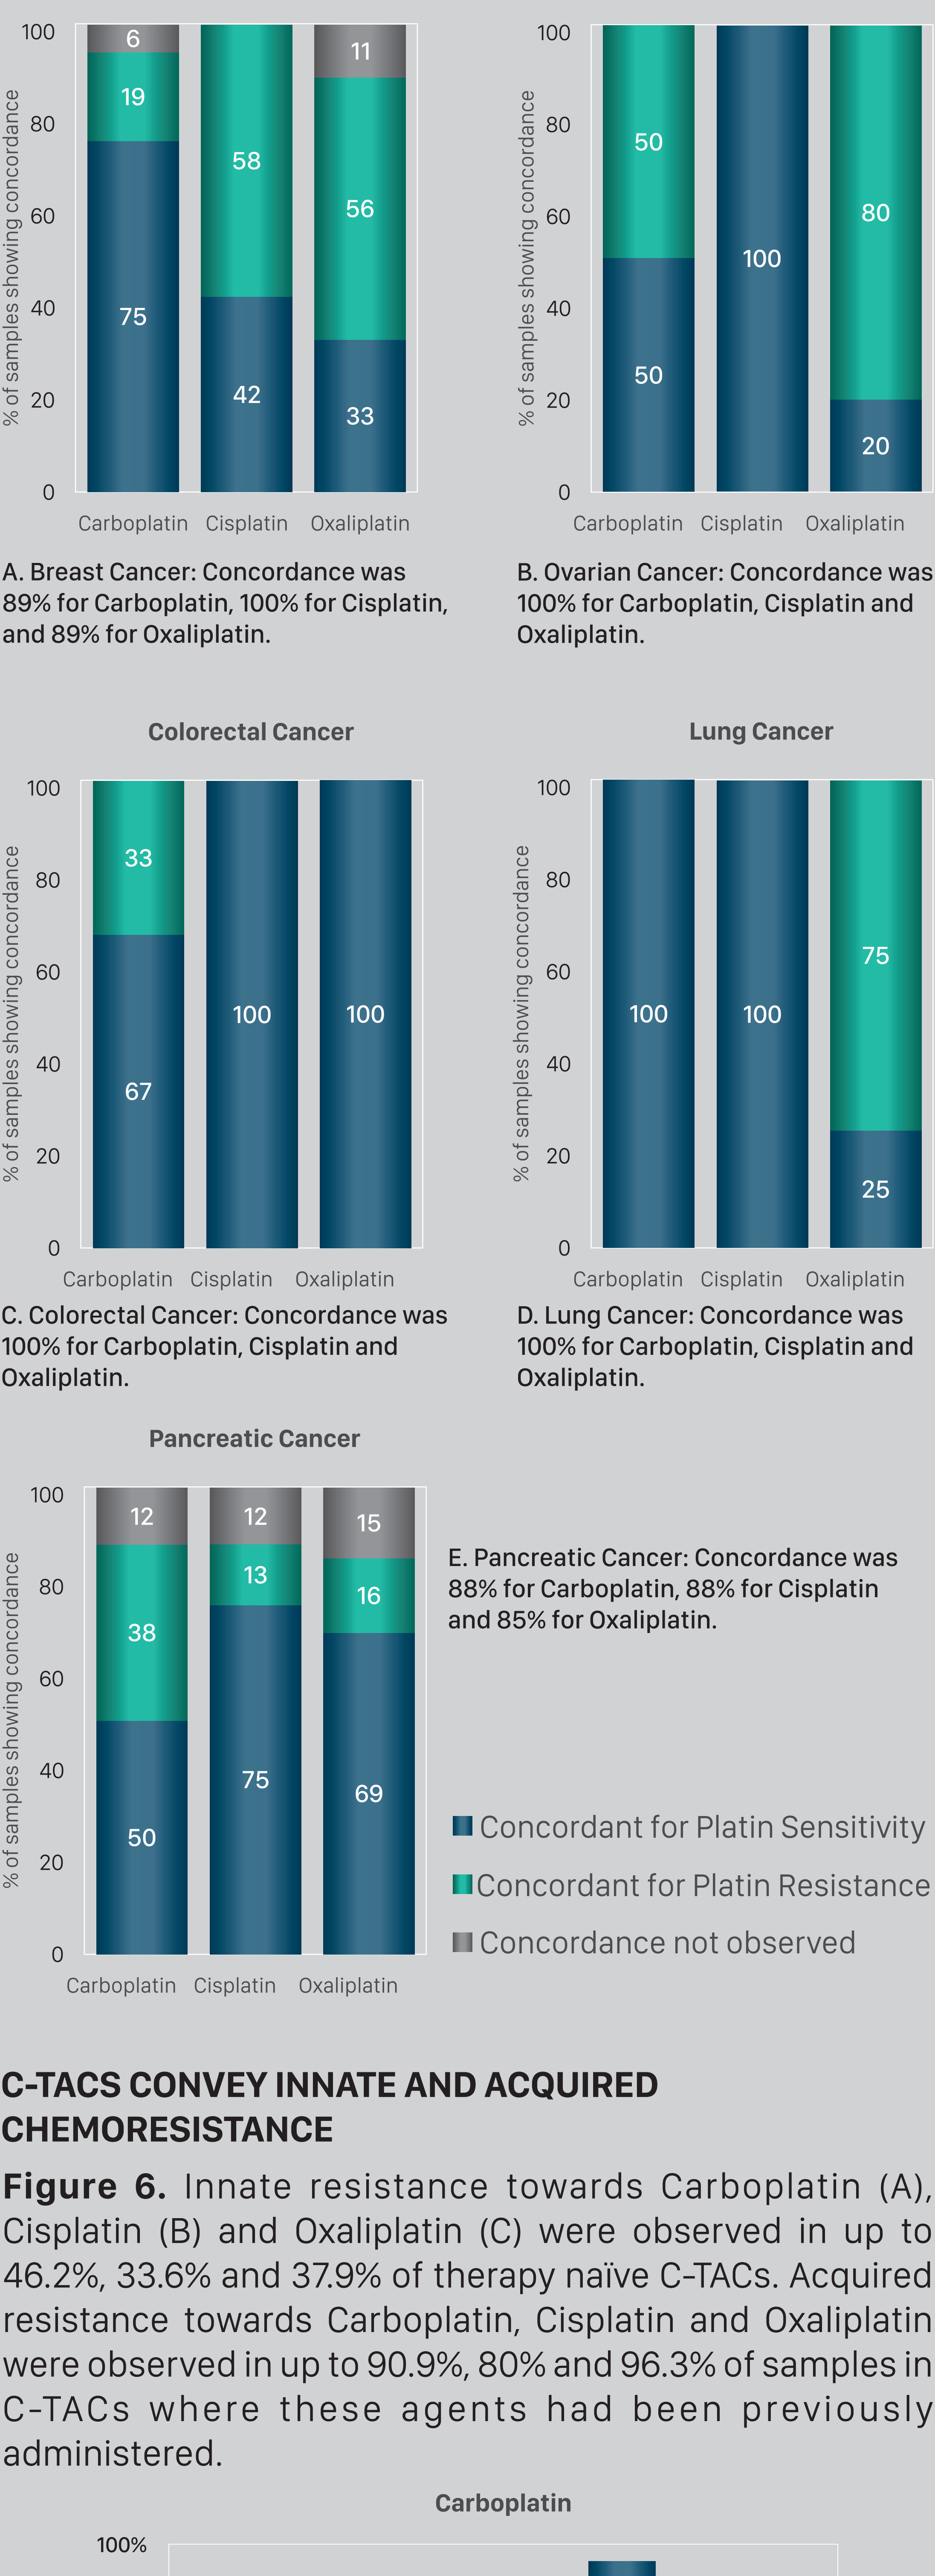
Breast Cancer
Ovarian Cancer
Colorectal Cancer
Lung Cancer
Pancreatic Cancer
% of samples showing concordance
Carboplatin
Cisplatin
Oxaliplatin
Concordant for Platin Sensitivity
Concordant for Platin Resistance
Concordance not observed

A. Breast Cancer: Concordance was 89% for Carboplatin, 100% for Cisplatin, and 89% for Oxaliplatin.

B. Ovarian Cancer: Concordance was 100% for Carboplatin, Cisplatin and Oxaliplatin.

C. Colorectal Cancer: Concordance was 100% for Carboplatin, Cisplatin and Oxaliplatin.

D. Lung Cancer: Concordance was 100% for Carboplatin, Cisplatin and Oxaliplatin.

E. Pancreatic Cancer: Concordance was 88% for Carboplatin, 88% for Cisplatin and 85% for Oxaliplatin.

C-TACS CONVEY INNATE AND ACQUIRED CHEMORESISTANCE

Figure 6. Innate resistance towards Carboplatin (A), Cisplatin (B) and Oxaliplatin (C) were observed in up to 46.2%, 33.6% and 37.9% of therapy naïve C-TACs. Acquired resistance towards Carboplatin, Cisplatin and Oxaliplatin were observed in up to 90.9%, 80% and 96.3% of samples in C-TACs where these agents had been previously administered.

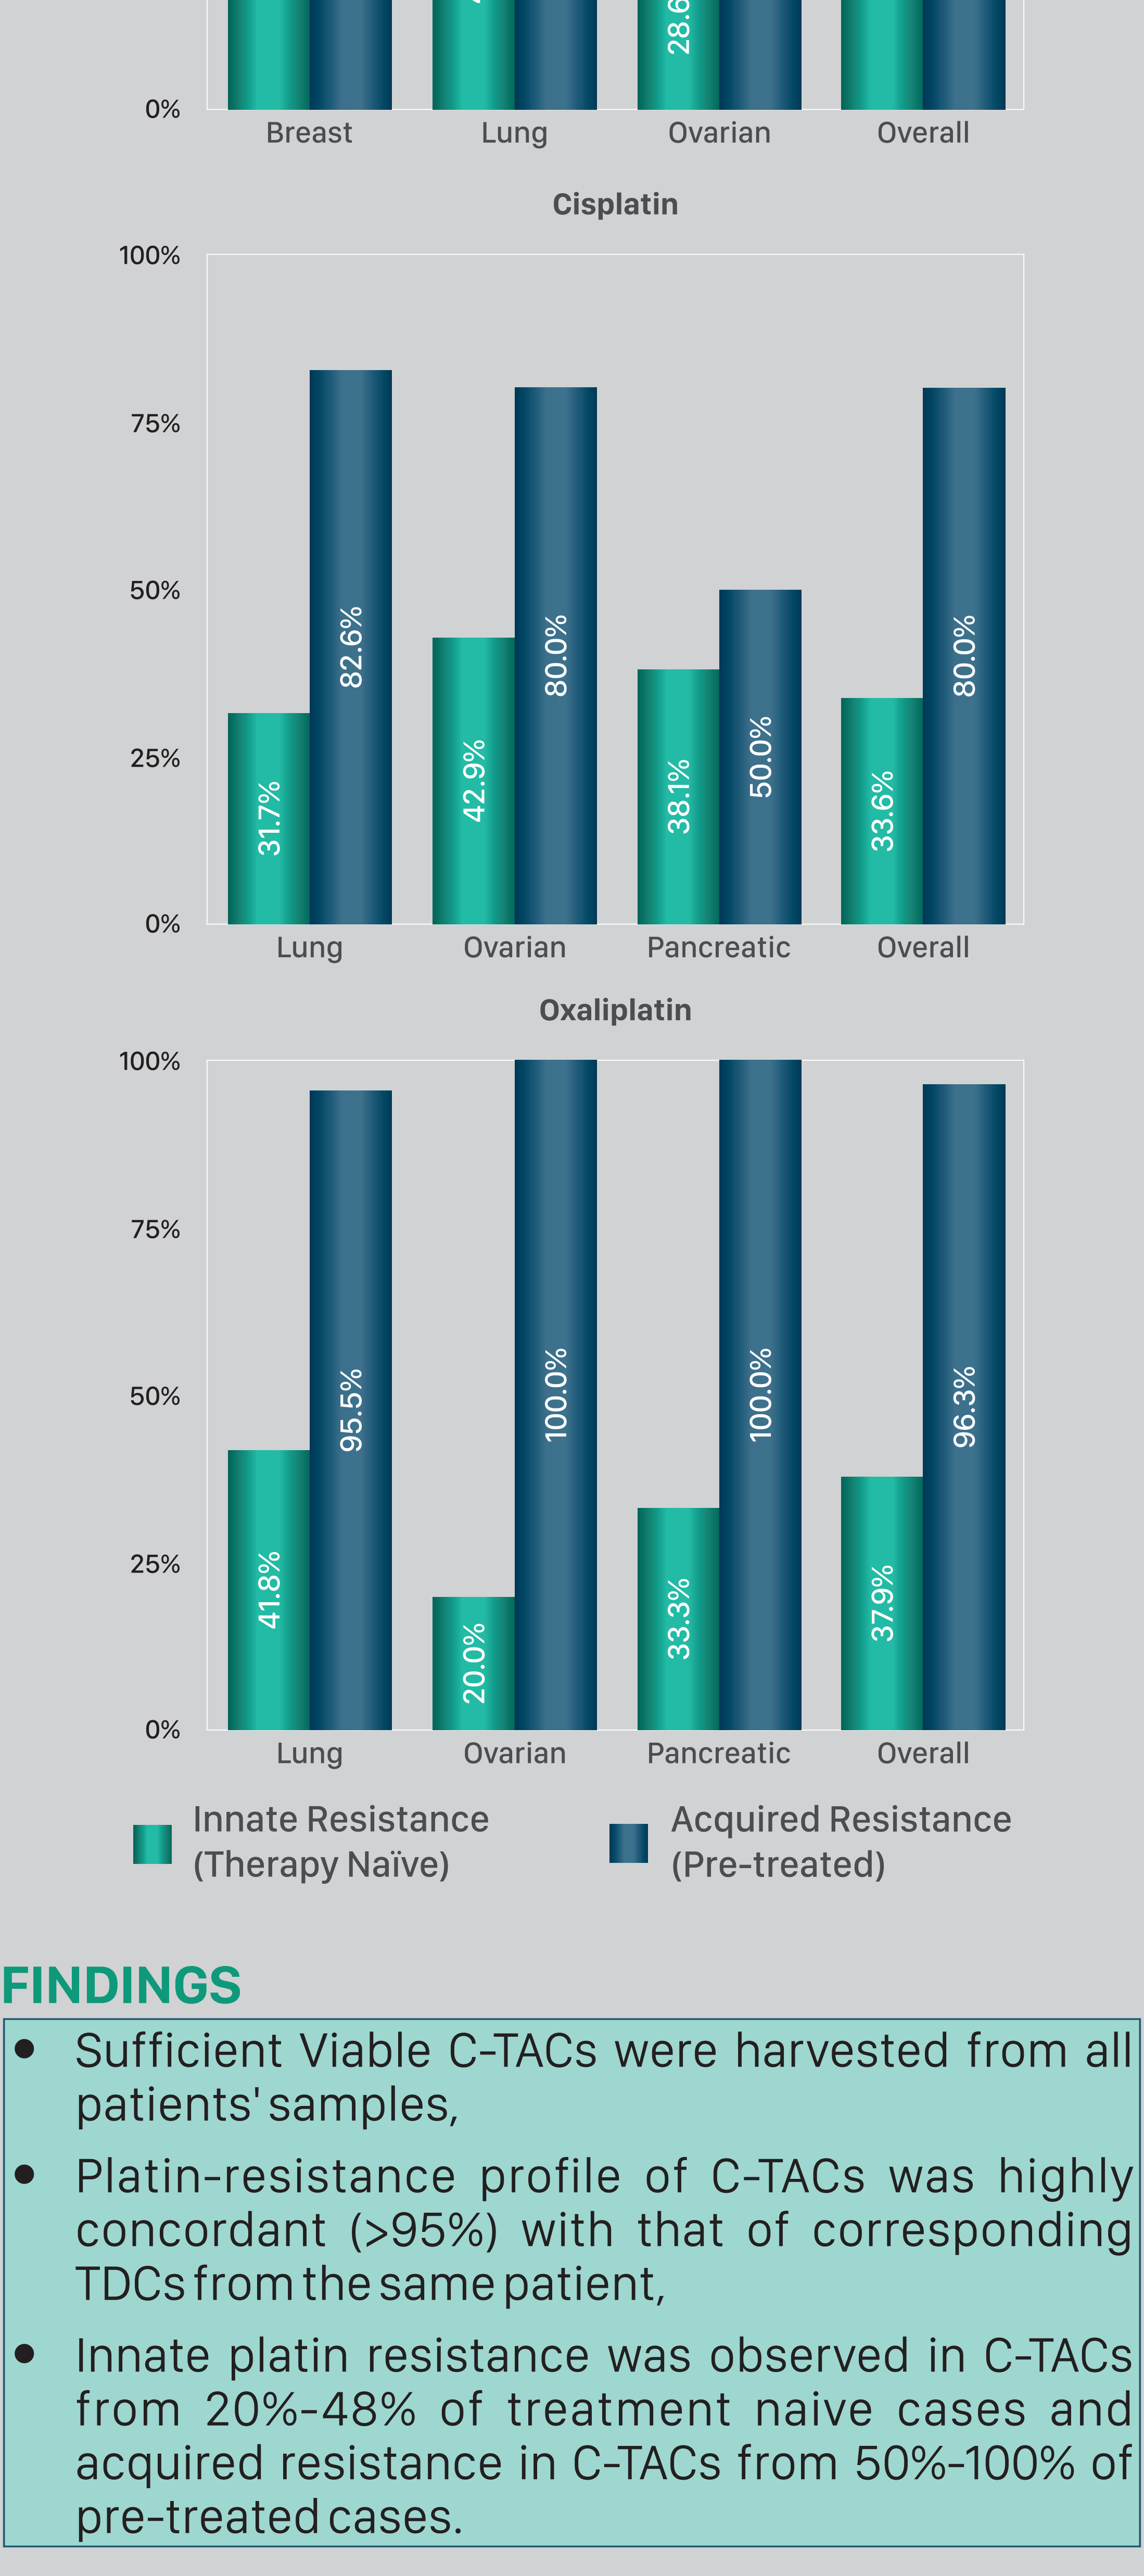
Carboplatin
Breast 48.1% 87.5%
Lung 43.9% 86.7%
Ovarian 28.6% 97.1%
Overall 46.2% 90.9%
Cisplatin
Lung 31.7% 82.6%
Ovarian 42.9% 80.0%
Pancreatic 38.1% 50.0%
Overall 33.6% 80.0%
Oxaliplatin
Lung 41.8% 95.5%
Ovarian 20.0% 100.0%
Pancreatic 33.3% 100.0%
Overall 37.9% 96.3%
Innate Resistance (Therapy Naïve)
Acquired Resistance (Pre-treated)

FINDINGS

- Sufficient Viable C-TACs were harvested from all patients' samples,
- Platin-resistance profile of C-TACs was highly concordant (>95%) with that of corresponding TDCs from the same patient,
- Innate platin resistance was observed in C-TACs from 20%-48% of treatment naive cases and acquired resistance in C-TACs from 50%-100% of pre-treated cases.

CONCLUSION

- It is possible to obtain sufficient viable C-TACs which accurately convey platin-resistance status.
- Chemoresistance profiling of C-TACs can be used for non-invasive, real-time monitoring of platin-resistance.
- C-TAC-based approach eliminates the need for painful invasive biopsies to obtain tumor tissue / cells.